CTX-8371, a Novel Bispecific Targeting both PD-1 and PD-L1, is more Potent than Combination anti-PD-1 and PD-L1 Therapy and Provides Enhanced Protection from Tumors *in vivo*

Diana Albu, Pearl Bakhru, Wilson Guzman, Michael Ophir, Rachel McCrory, Beau Carson, Amanda Oliphant, Rachel Rennard, Lucy Liu, Alan Leung, Sara Haserlat, Michael Schmidt, Jose Gonzalo, Bing Gong, Robert Tighe, Ben Wolf

Compass Therapeutics, 245 First Street, Cambridge MA

Abstract

Background Monoclonal antibody immunotherapies targeting immune checkpoint receptors have shown great promise for a subset of cancer patients. However, novel combination therapies are still needed to increase the benefit of cancer immunotherapy and bring it to broader patient populations. Here we describe the preclinical evaluation of CTX-8371, which combines PD-1 and PD-L1 targeting in one bispecific, tetravalent molecule.

Methods Our proprietary Stitchmabs™ bispecific screening platform was used to conduct an unbiased screen of bispecifics comprised of various checkpoint blocking antibodies. This screen yielded the surprising discovery that a bispecific containing both PD-1 and PD-L1 binding arms was more potent than the combination of parent monoclonal antibodies. We then generated common light chain bispecifics containing compatible anti-PD-1 and PD-L1 antibodies and used multiple *in vitro* assays to identify our lead, CTX-8371. Additional *in vitro* and *in vivo* experiments confirmed CTX-8371's reactivity across species, *in vivo* anti-tumor effects, and the underlining mechanisms driving its distinctive activity.

Results We found that CTX-8371 binds to human and cynomolgus monkey PD-1 and PD-L1 targets with sub-nanomolar affinities and is cross-reactive to mouse PD-1 and PD-L1. Compared to Keytruda®, CTX-8371 increased T cell activation and tumor cell killing *in vitro*, significantly delayed tumor growth, and prolonged survival in human cell transfer tumor models. Additionally, CTX-8371 demonstrated efficacy in transplantable mouse syngeneic models. Investigation into the mechanisms responsible for the enhanced efficacy of CTX-8371 unexpectedly found that the bispecific causes a massive loss of PD-1 from the T cell surface, which was not observed in response to monoclonal antibodies alone or combined. This robust PD-1 downregulation, potentially mediated through bridging together the T cell and tumor cell, may explain the ability of CTX-8371 to reverse PD-1 suppression more potently than standard blocking antibodies.

Conclusions Taken together, the results demonstrate that the bispecific, tetravalent antibody CTX-8371 has increased potency *in vitro* and *in vivo* as compared to clinical checkpoint blockade agents. Some of its effects are likely attributable to its unique mechanism of action, driving robust downregulation of cell-surface PD-1. Thus, CTX-8371 has the potential to increase the number of patients that benefit from PD-1/PD-L1 checkpoint blockade.

Discovery of PD-1xPD-L1 synergy via a Stitchmab™ experiment

Stitchmabs allow for rapid testing of bispecific concepts

- We initially theorized that a bispecific containing anti-PD-1 and PD-L1 arms would function to bring T cells into contact with PD-L1+ tumor cells while also blocking a critical T cell checkpoint

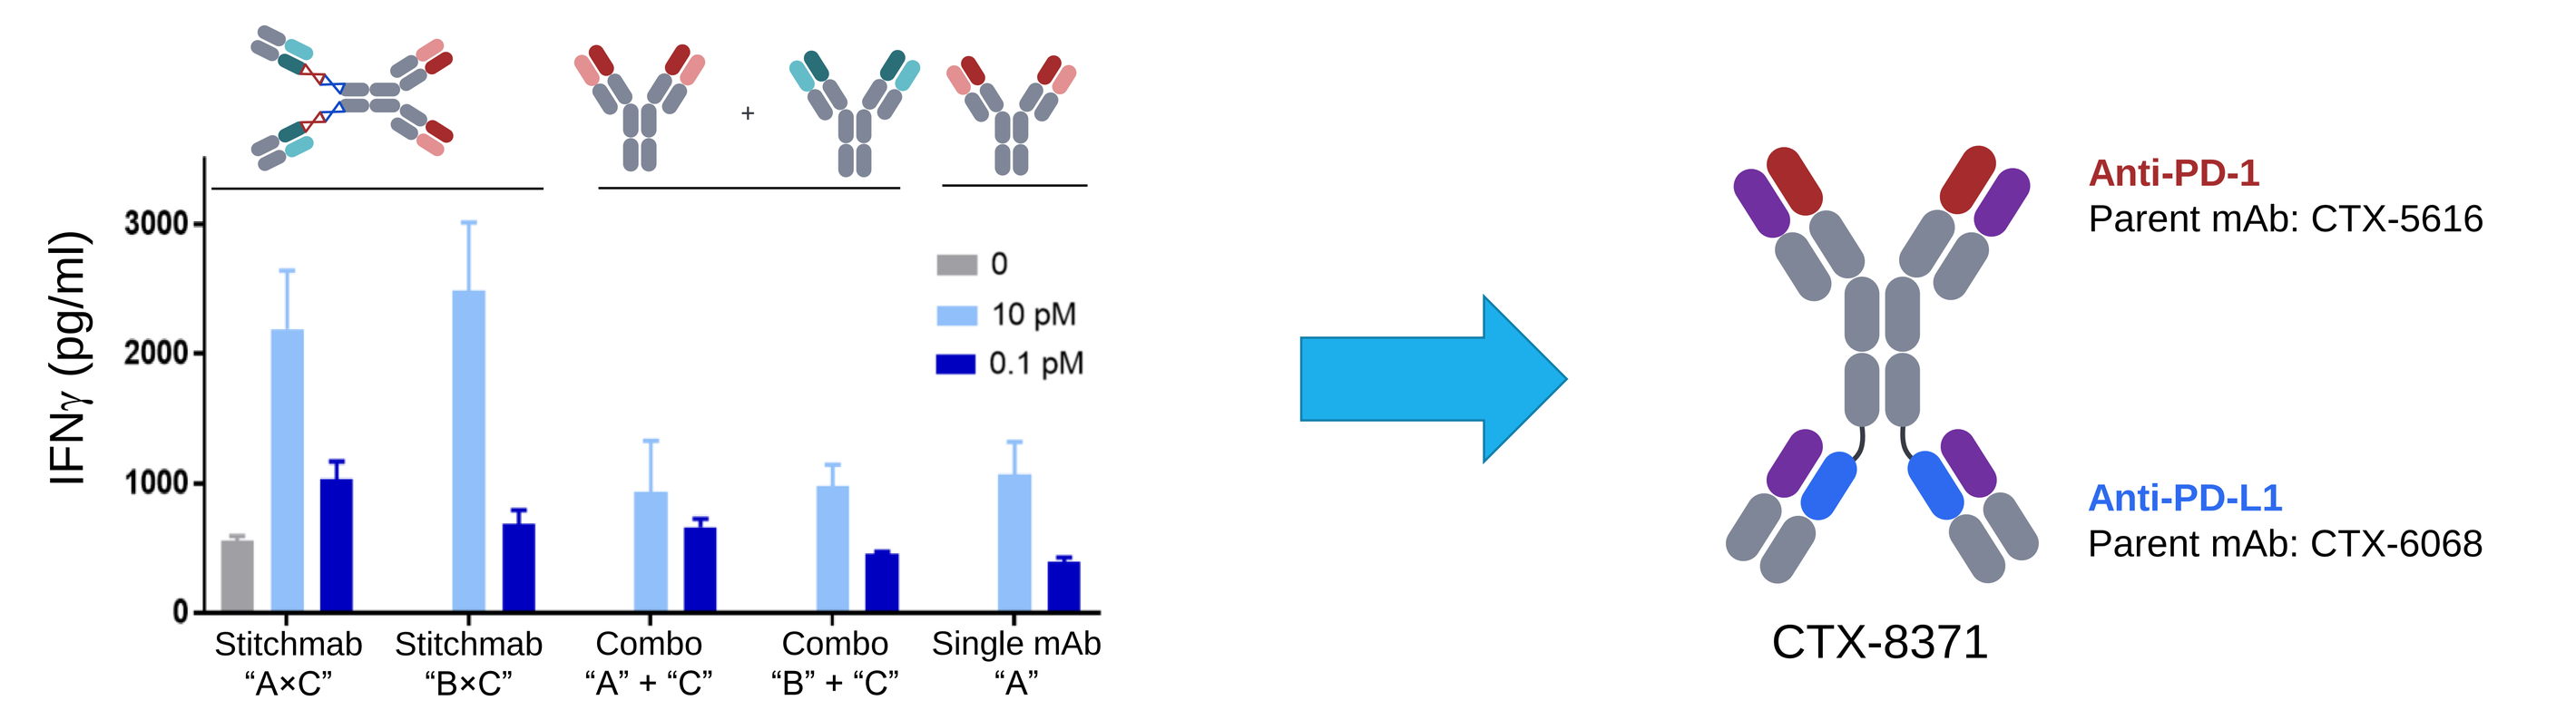
IFNγ (pg/ml)
3000
2000
1000
0
0
10 pM
0.1 pM
Stitchmab "A×C"
Stitchmab "B×C"
Combo "A" + "C"
Combo "B" + "C"
Single mAb "A"
Anti-PD-1
Parent mAb: CTX-5616
Anti-PD-L1
Parent mAb: CTX-6068
CTX-8371

- In a MLR assay, Stitchmab™ bispecifics of commercial anti-PD-1 and anti-PD-L1 antibodies caused increased IFN-γ production at picomolar antibody levels
 - This prompted us to generate common light chain bispecific variants
- CTX-8371 was selected as a PD-1×PD-L1 bispecific from in-house generated anti-PD-1 and anti-PD-L1 antibodies with excellent drug-like properties

Affinity (nM)	Human PD-1	Cyno PD-1
CTX-5616 (a-PD-1)	1.09e-09	1.97e-09
Keytruda	1.81e-09	7.42e-10
Nivolumab	3.52e-09	1.34e-09

Affinity (nM)	Human PD-L1	Cyno PD-L1
CTX-6068 (a-PD-L1)	3.08e-09	2.30e-09
Atezolizumab	4.55e-10	1.05e-08
Avelumab	5.05e-10	7.51e-10

- Compass anti-PD-1 and anti-PD-L1 antibody Fabs have similar affinities as clinical comparator Fabs to Human and Cynomolgus monkey PD-1 and PD-L1

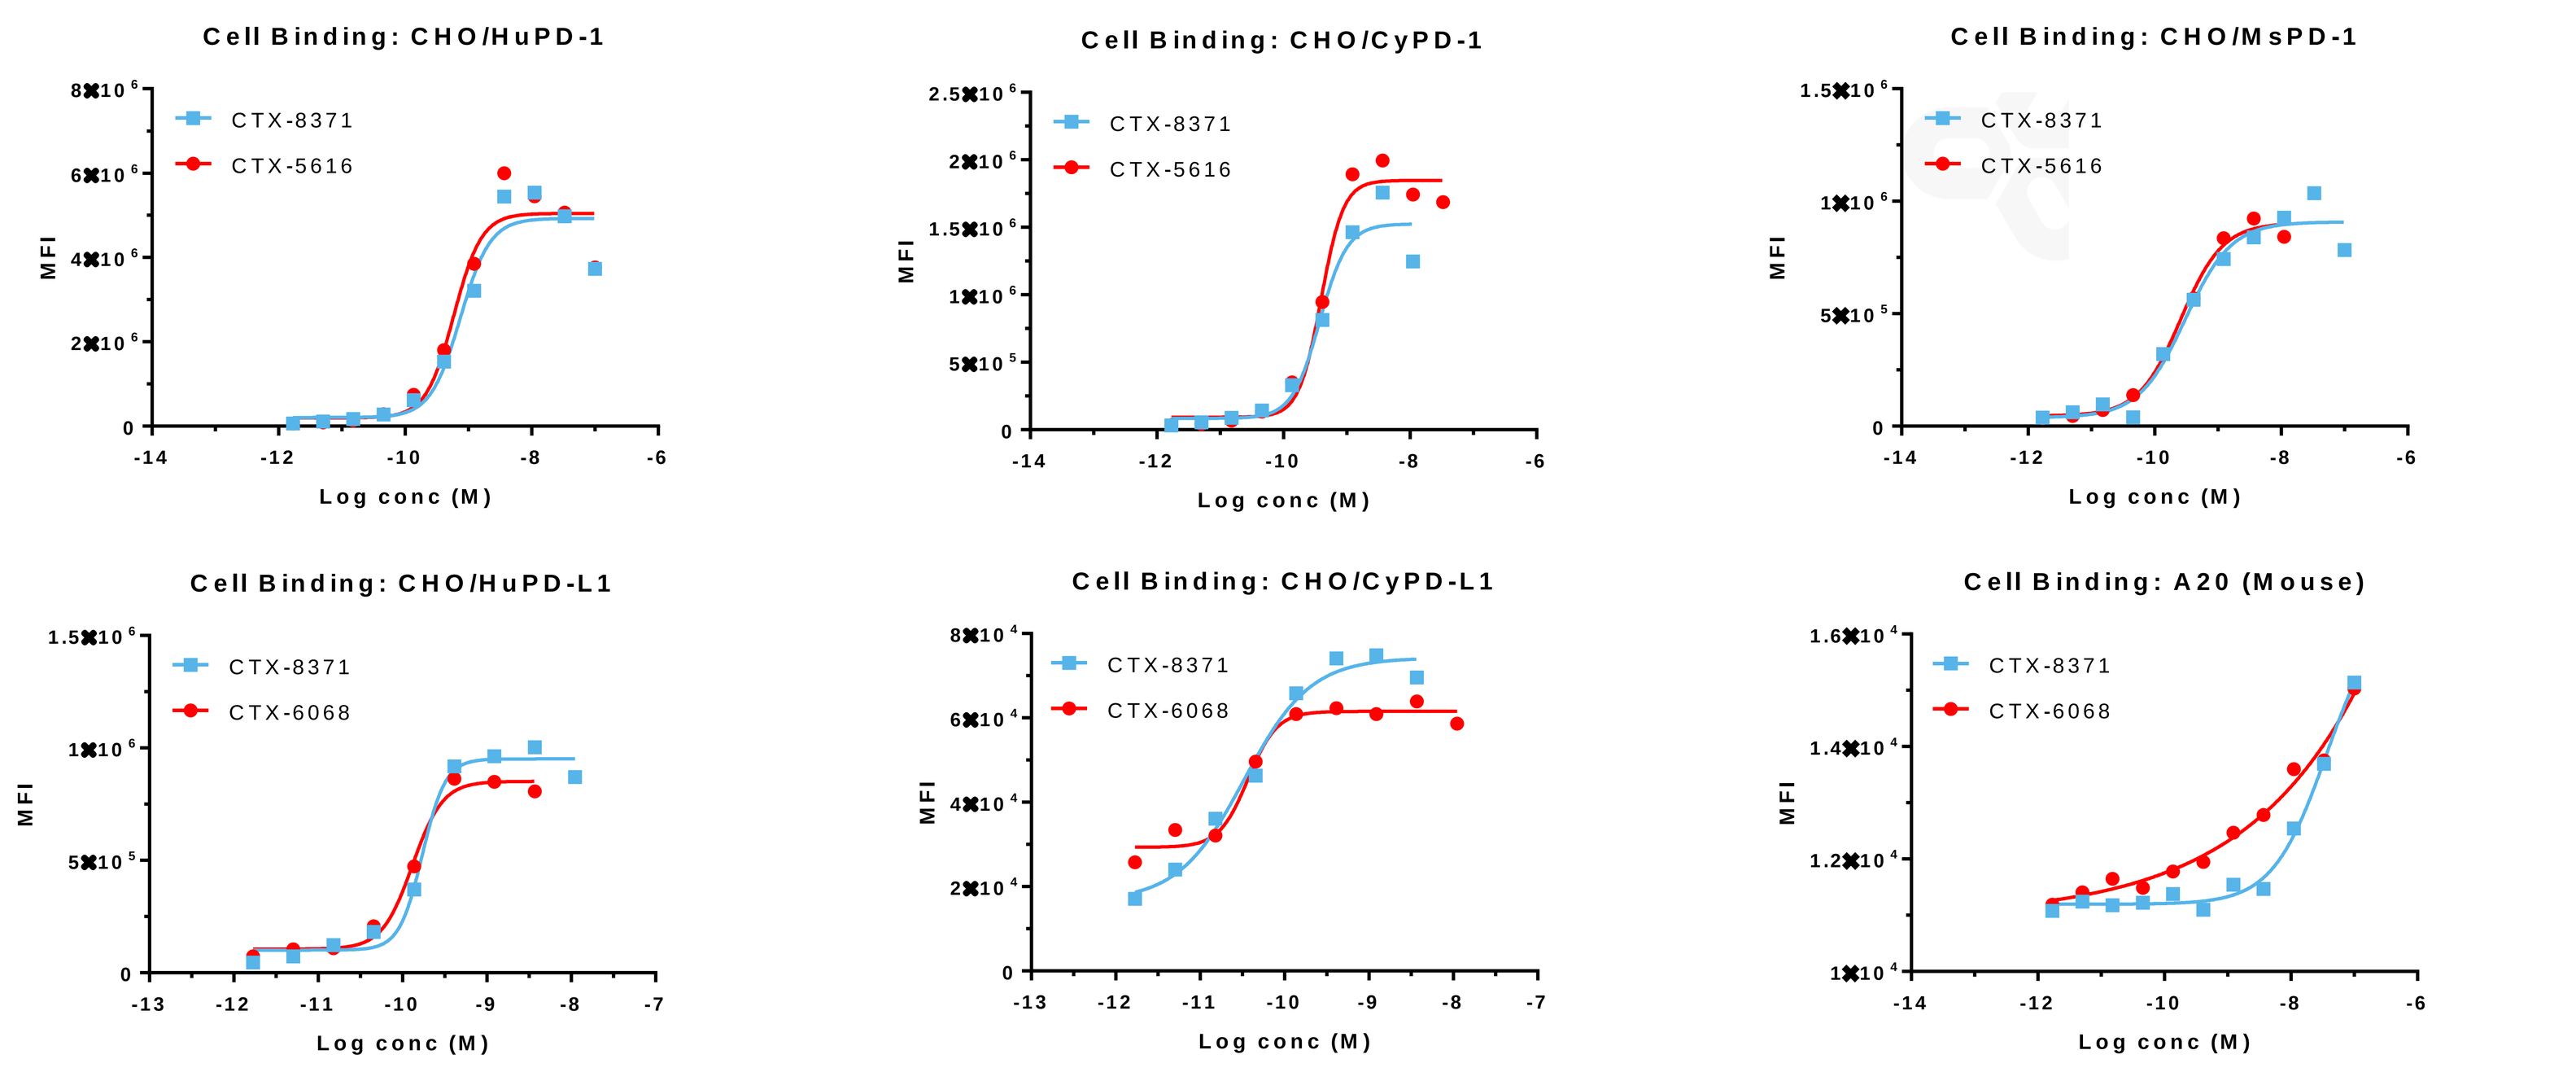
Cell Binding: CHO/HuPD-1
Cell Binding: CHO/CyPD-1
Cell Binding: CHO/MsPD-1
Cell Binding: CHO/HuPD-L1
Cell Binding: CHO/CyPD-L1
Cell Binding: A20 (Mouse)
CTX-8371
CTX-5616
CTX-6068
MFI
Log conc (M)

- CTX-8371 maintains monoclonal antibody-like binding to PD-1 and PD-L1 from all 3 species as compared to monoclonal parent molecules.
 - CTX-8371 also has strong anti-mouse PD-1 binding but weaker anti-mouse PD-L1 binding

CTX-8371 increases T cell activation and killing *in vitro*

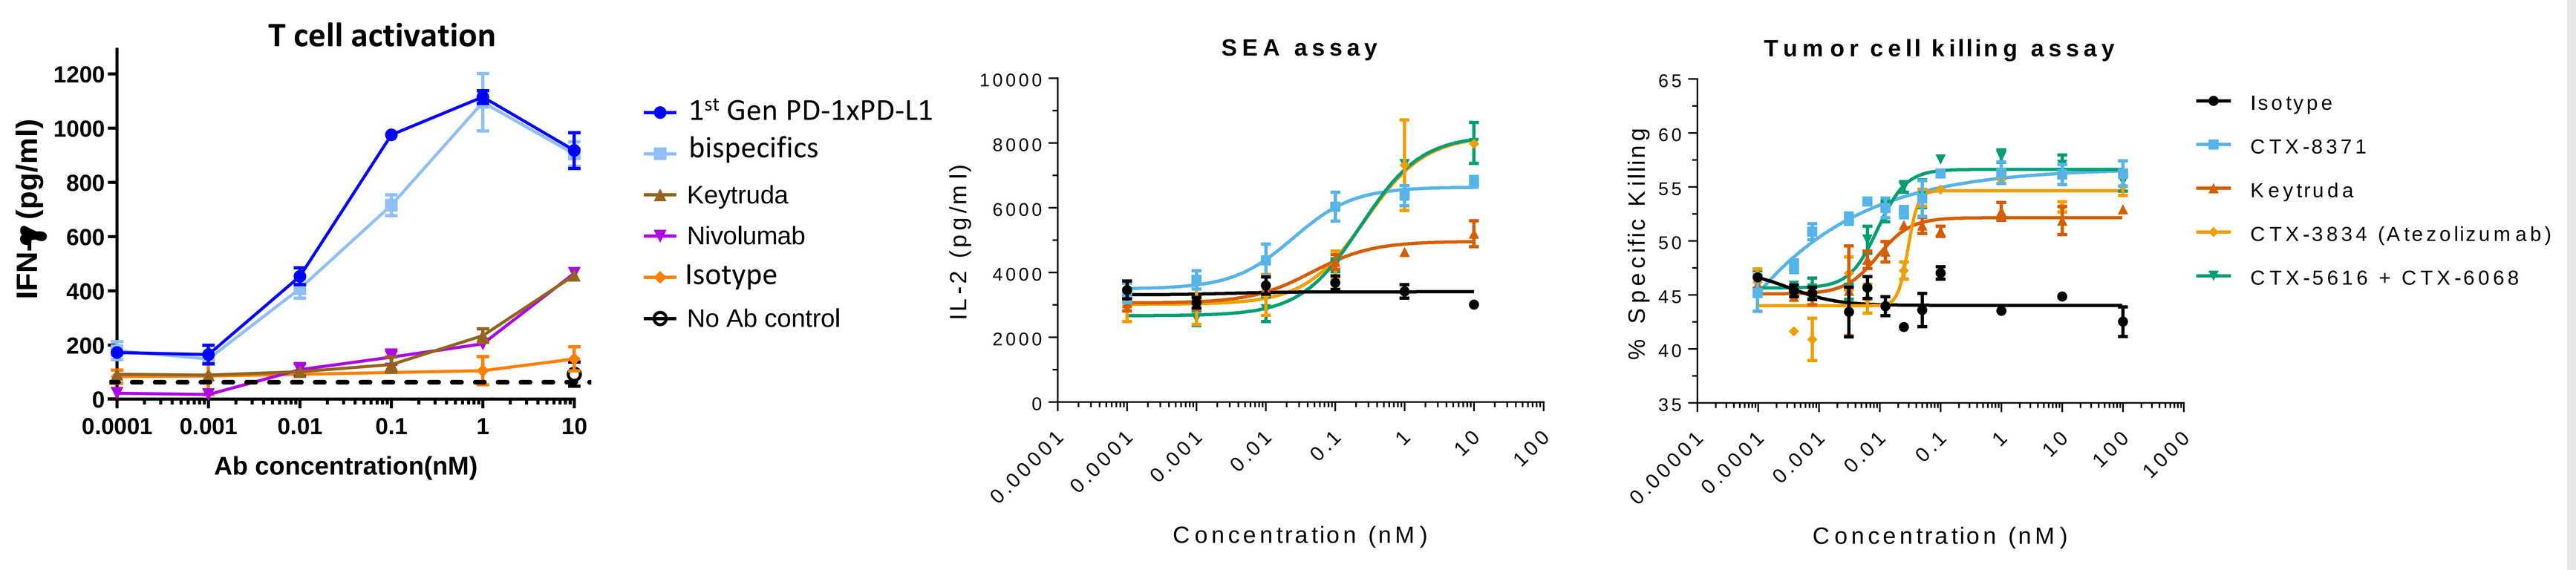
T cell activation
IFNγ (pg/ml)
Ab concentration(nM)
1st Gen PD-1xPD-L1 bispecifics
Keytruda
Nivolumab
Isotype
No Ab control
SEA assay
IL-2 (pg/ml)
Concentration (nM)
Tumor cell killing assay
% Specific Killing
Concentration (nM)
Isotype
CTX-8371
Keytruda
CTX-3834 (Atezolizumab)
CTX-5616 + CTX-6068

- Human T cells were expanded *in vitro* by TCR activation for 7 days and co-cultured with engineered K562 tumor cell targets in the presence of antibodies for 2 days
 - 1st generation PD-1xPD-L1 bispecifics enhanced T cell activation compared to Keytruda, Nivolumab, and controls
- Human PBMCs were stimulated with Staphylococcus Enterotoxin A (SEA) and treated with CTX-8371 or other antibodies for 3 days
 - CTX-8371 remained active at sub-nanomolar doses while other antibodies have regressed to the isotype
- CMV-specific T cells were co-cultured with engineered K562 tumor cell targets and bystander cells in the presence of antibodies for 2 days
 - CTX-8371 treatment increased CMV-specific T cell killing of K562 tumor cell targets at sub-nanomolar doses while the other antibodies regressed to the isotype

Unique MOA: CTX-8371 drives loss of PD-1

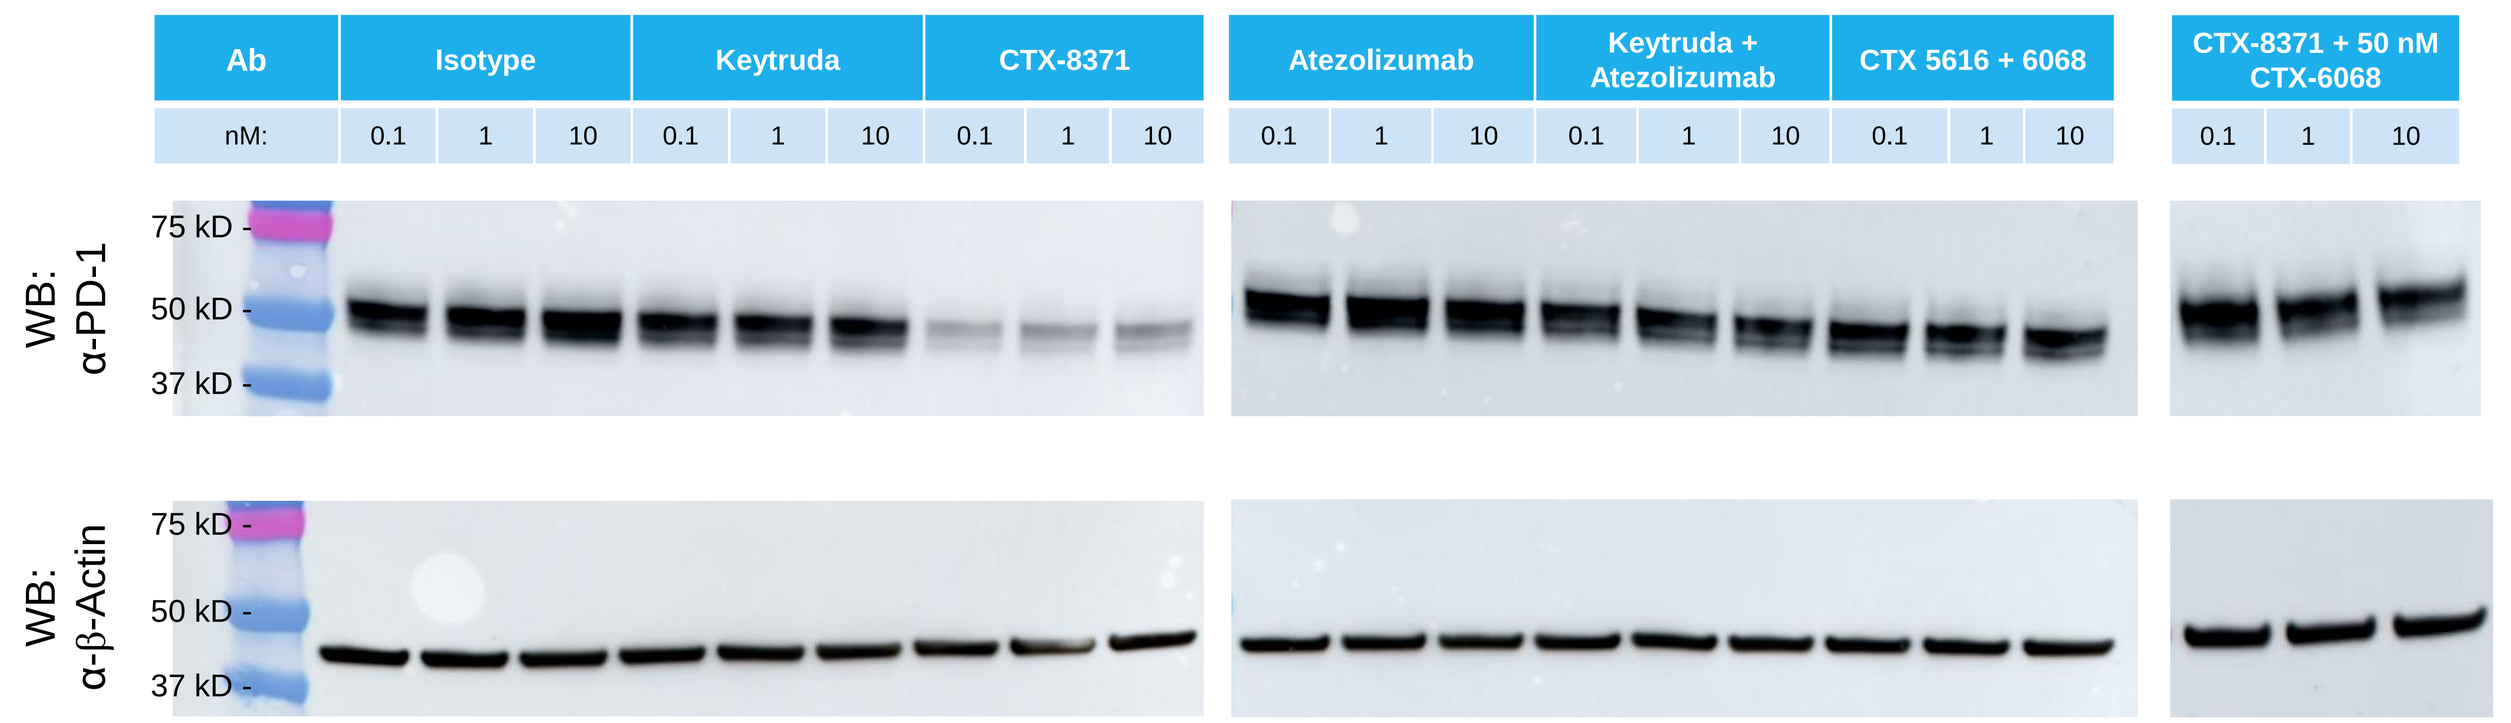
Ab
Isotype
Keytruda
CTX-8371
Atezolizumab
Keytruda + Atezolizumab
CTX 5616 + 6068
CTX-8371 + 50 nM CTX-6068
nM:
0.1
1
10
WB: α-PD-1
WB: α-β-Actin
75 kD
50 kD
37 kD

- Human PBMCs were stimulated with anti-CD3 and anti-CD28 antibodies for 3 days before overnight co-culture with the listed antibodies
 - CTX-8371 drove loss of cell-associated PD-1 but only when both arms of the bispecific were engaged

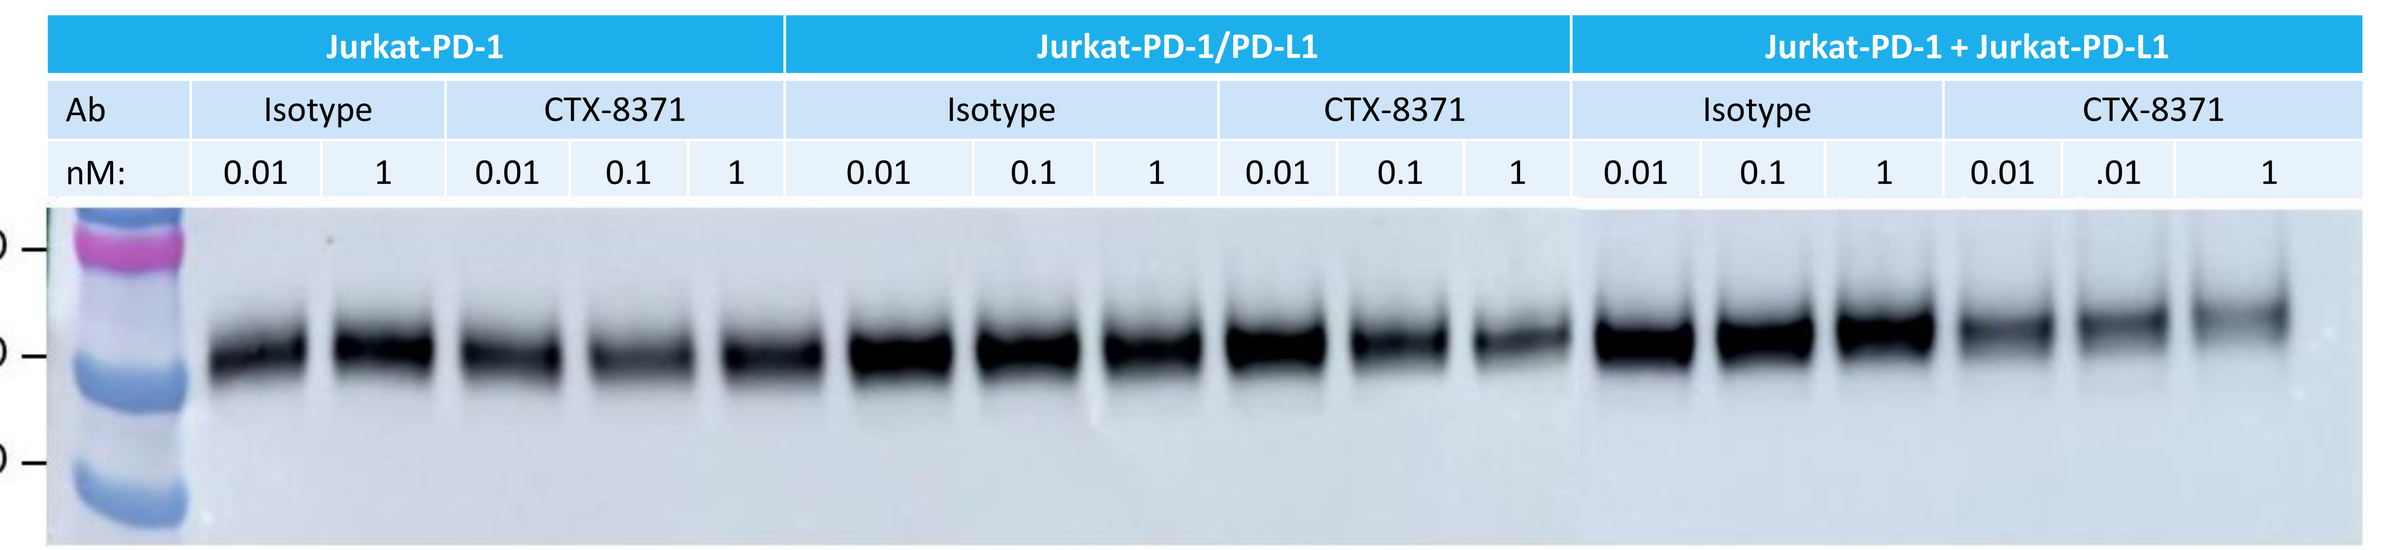
Jurkat-PD-1
Jurkat-PD-1/PD-L1
Jurkat-PD-1 + Jurkat-PD-L1
Ab
Isotype
CTX-8371
nM:
0.01
0.1
1
75kD
50kD
37kD

- Jurkat-PD1, Jurkat-PD-1/PD-L1, or mix of Jurkat-PD-1 + Jurkat-PD-L1 were treated overnight with isotype or CTX-8371 antibodies
 - PD-L1 binding was necessary for CTX-8371-induced loss of cell-associated PD-1 (when both arms of the bispecific were engaged)
 - CTX-8371 drove PD-1 loss predominantly when PD-1 and PD-L1 were in trans

CTX-8371 is effective against mouse syngeneic tumors

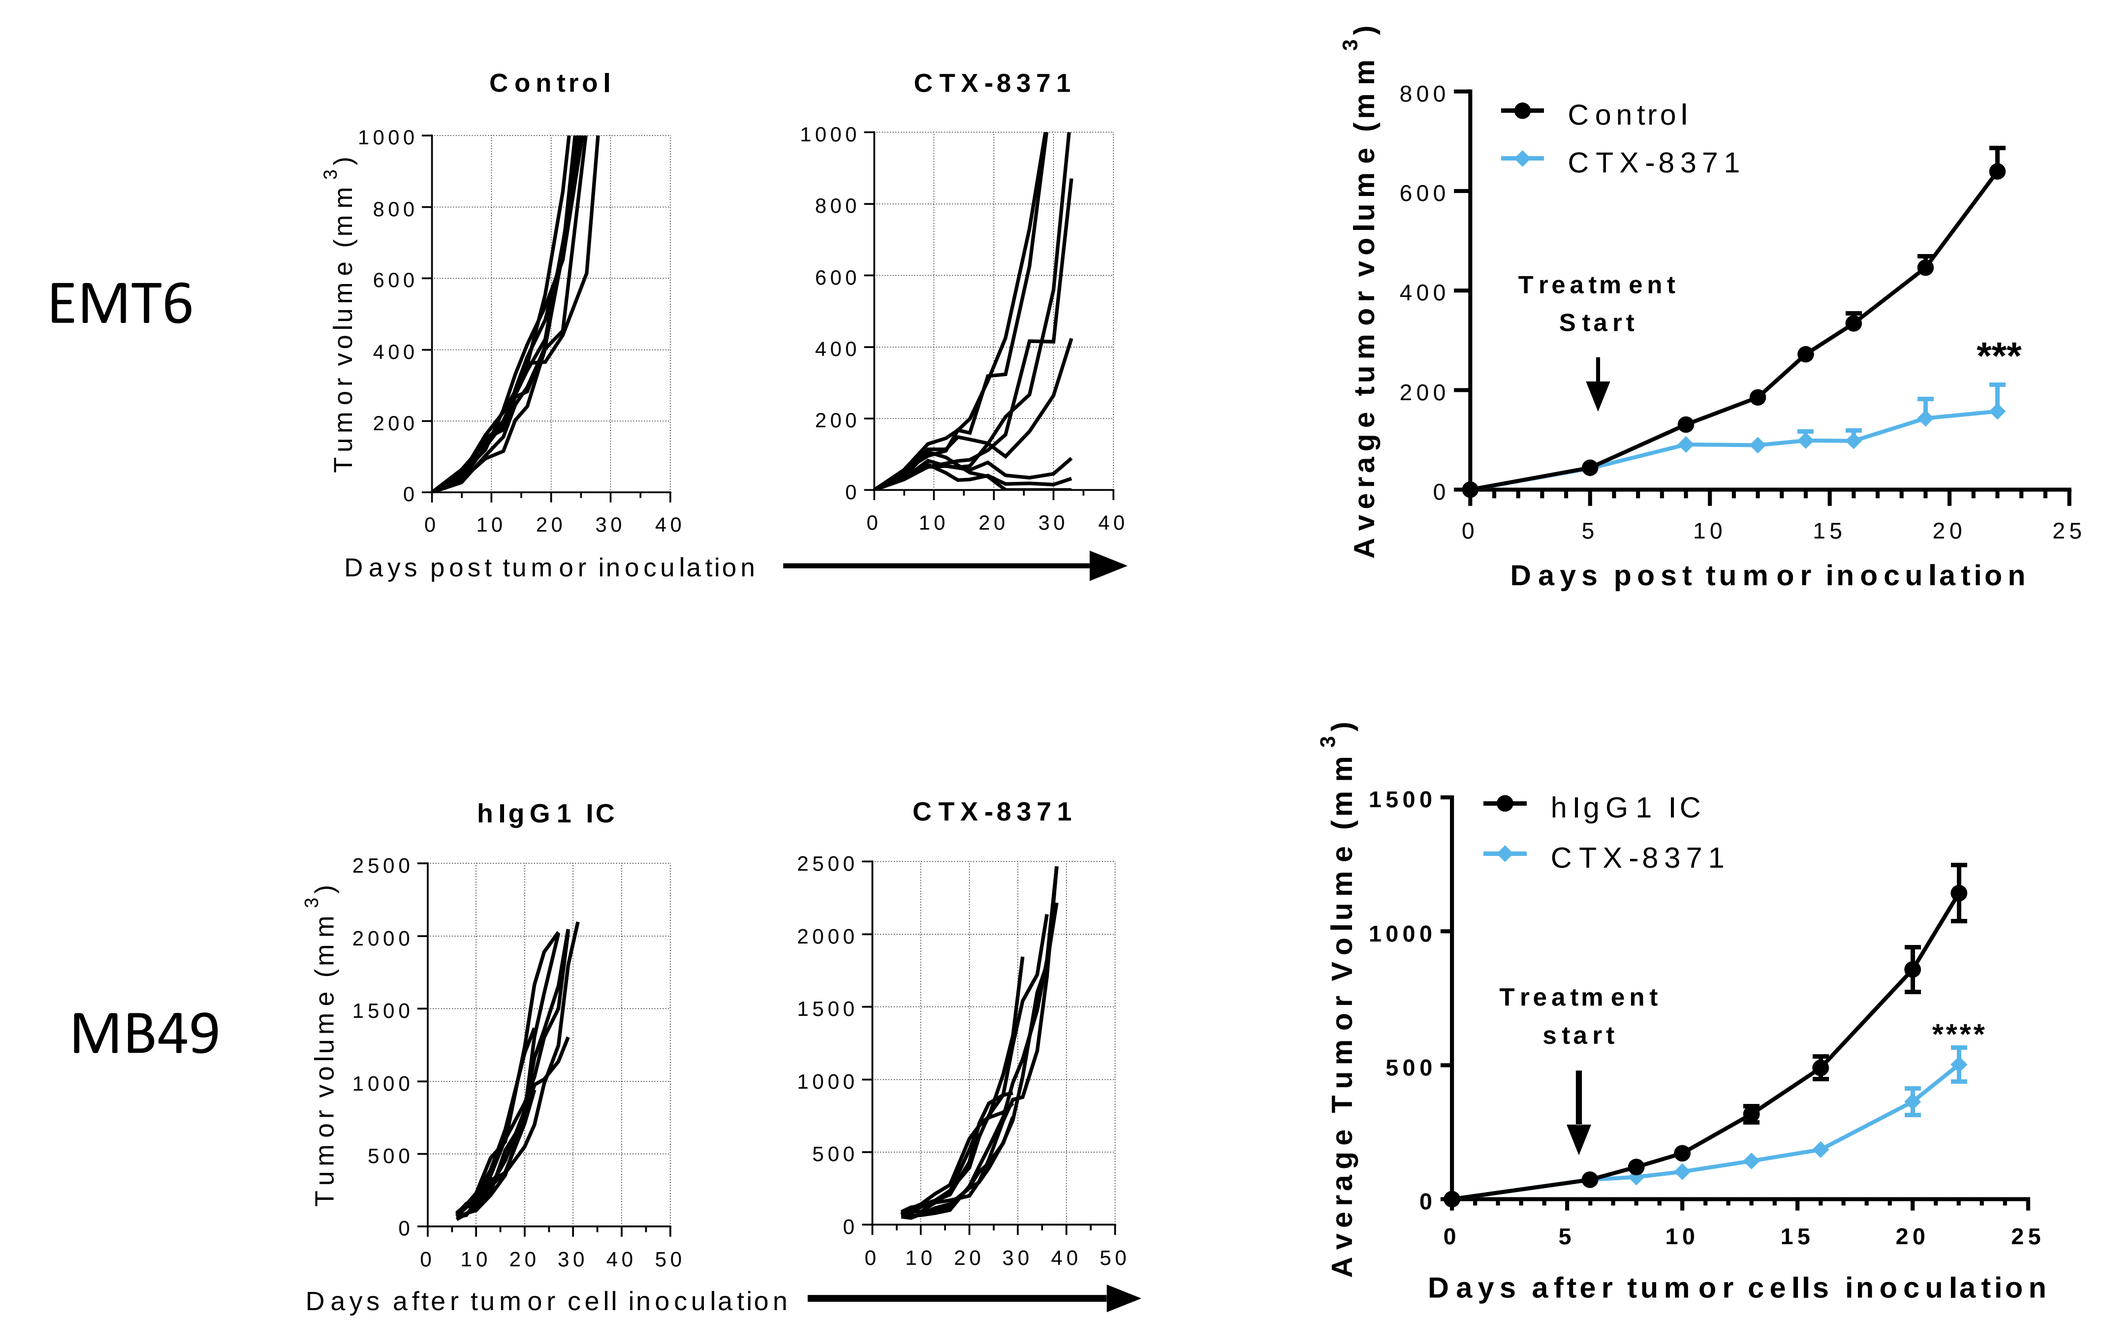
EMT6
Control
CTX-8371
Tumor volume (mm3)
Days post tumor inoculation
Average tumor volume (mm3)
Treatment Start

MB49
hIgG1 IC
CTX-8371
Days after tumor cell inoculation
Average Tumor Volume (mm3)
Treatment start

Days after tumor cells inoculation

- CTX-8371 is effective against mouse syngeneic tumors, although its potency in syngeneic tumors might be underestimated due to the very weak mouse PD-L1 cross-reactivity
 - Upper panel shows individual and average tumor growth curves for EMT-6 mouse breast cancer treated with CTX-8371 or isotype control antibodies
 - Lower panel shows individual and average tumor growth curves for MB49 mouse bladder carcinoma treated with CTX-8371 or isotype control antibodies
 - ****, P<0.0001, ***, P<0.001, Two-way ANOVA and Tukey's multiple comparisons test.

CTX-8371 has potent anti-tumor activity against mouse tumors implanted in PD-1/PD-L1 humanized mice with no apparent toxicity

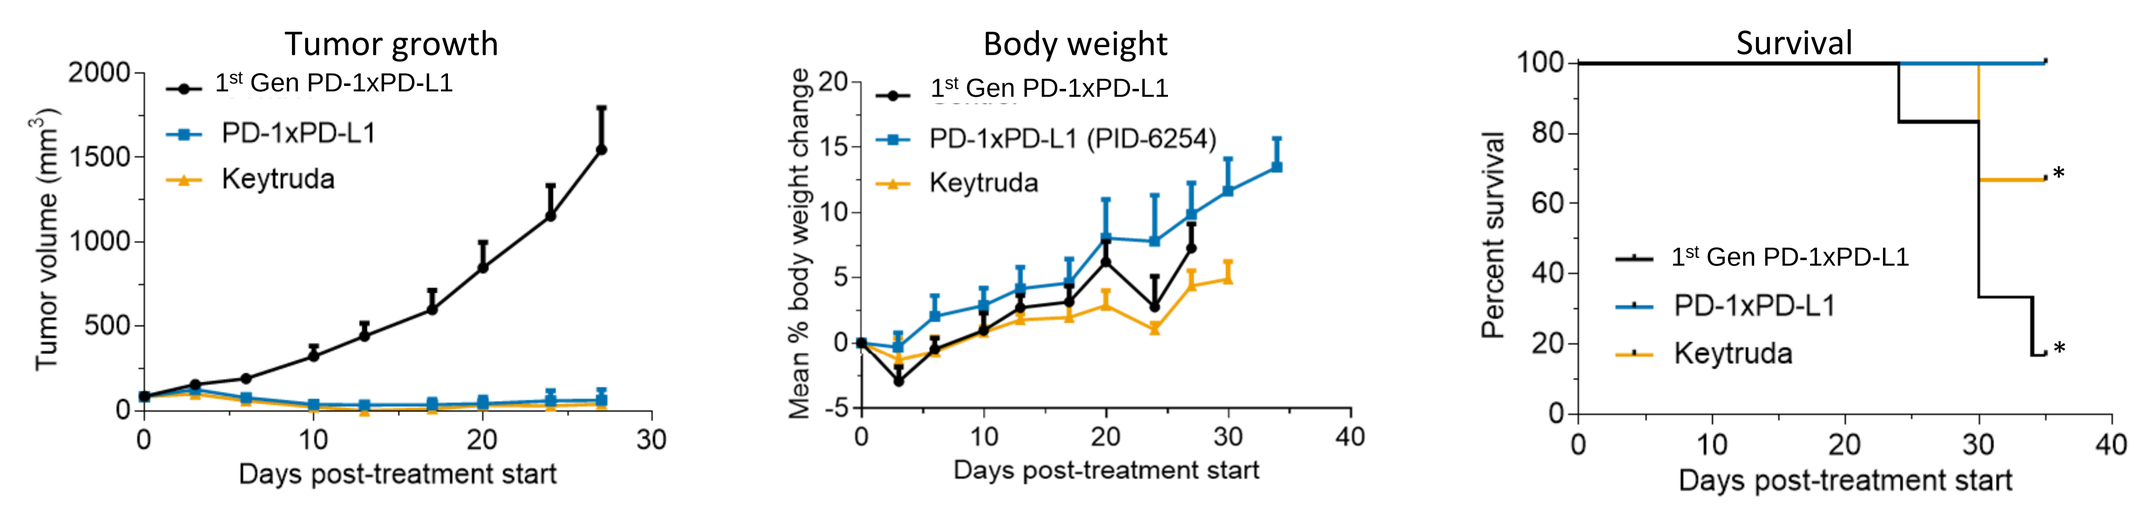
Tumor growth
Body weight
Survival
1st Gen PD-1xPD-L1
PD-1xPD-L1
PD-1xPD-L1 (PID-6254)
Keytruda
Tumor volume (mm3)
Mean % body weight change
Percent survival
Days post-treatment start

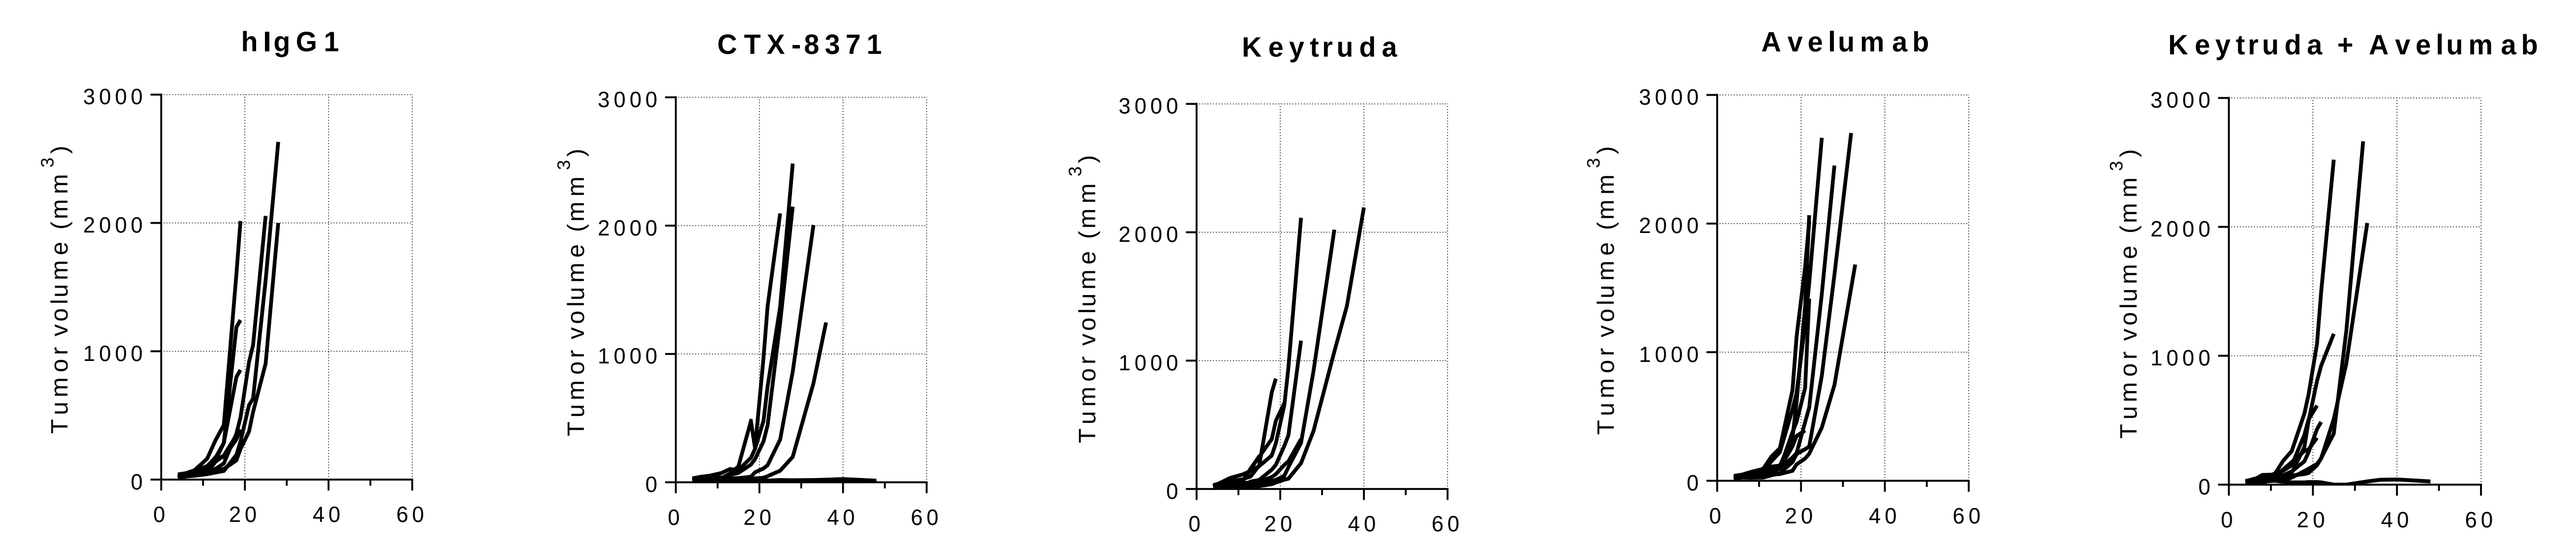
hIgG1
CTX-8371
Keytruda
Avelumab
Keytruda + Avelumab
Tumor volume (mm3)

Treatment	Tumor-free/Total
IC hIgG1	0/8
CTX-8371	3/8
Keytruda	0/8
Avelumab	0/8
Keytruda + Avelumab	1/8

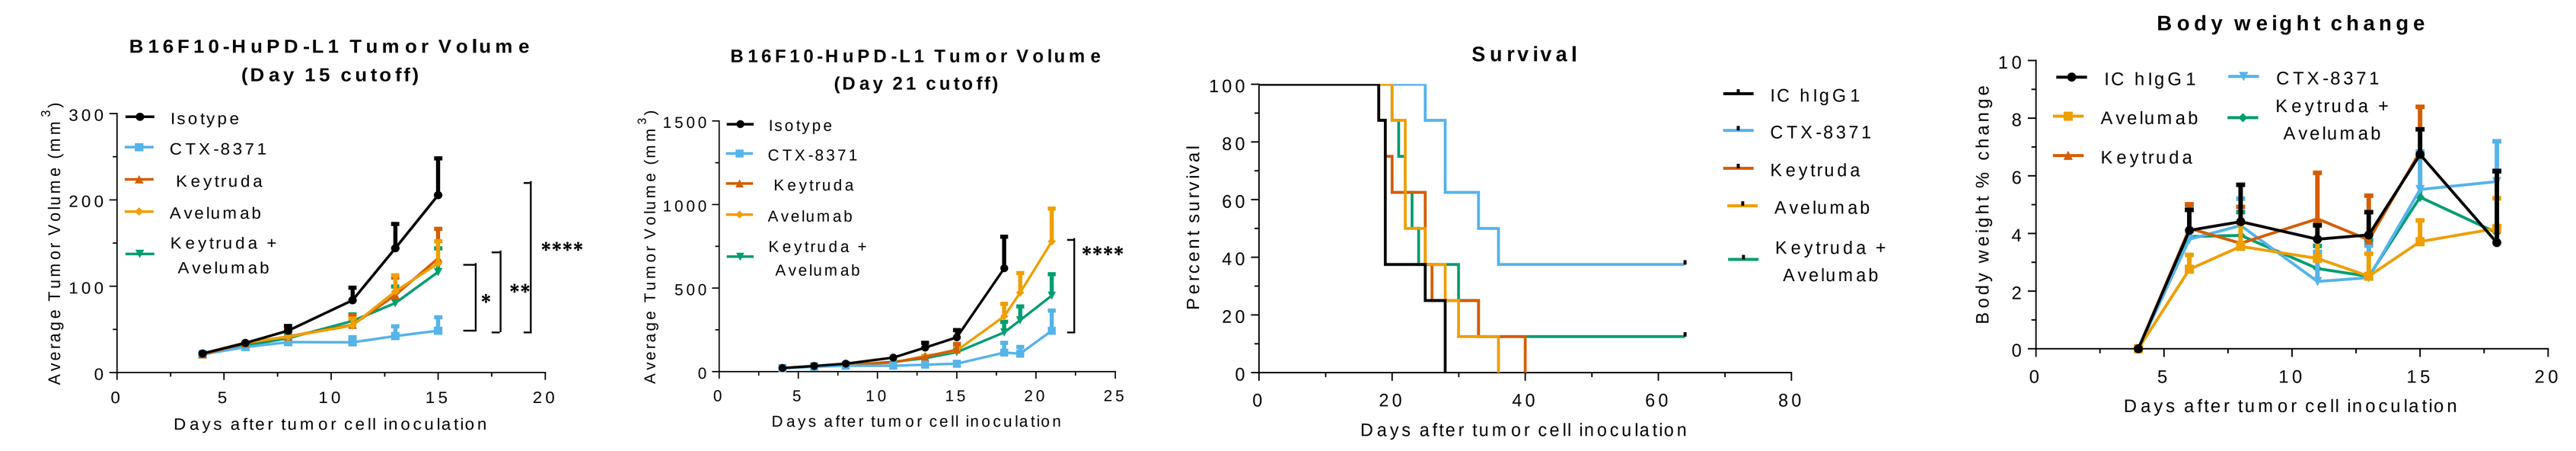
B16F10-HuPD-L1 Tumor Volume (Day 15 cutoff)
B16F10-HuPD-L1 Tumor Volume (Day 21 cutoff)
Survival
Body weight change
Isotype
IC hIgG1
CTX-8371
Keytruda
Avelumab
Keytruda + Avelumab
Average Tumor Volume (mm3)
Percent survival
Body weight % change
Days after tumor cell inoculation

- CTX-8371 is effective against mouse tumors expressing human PD-L1
 - Upper graphs show average tumor growth curves, body weight, and survival of huPD-1/PD-L1 transgenic mice bearing MC-38-hPD-L1 tumors. * 2 mice in Keytruda group and 1 mouse in control group were euthanized due to dermatitis
 - Lower graphs depict individual and average tumor growth curves, survival, and body weight change of transgenic C57BL/6-huPD-1/PD-L1 mice bearing B16-F10-huPD-L1 melanomas. Different groups of mice (n=8) were treated with CTX-8371, Keytruda, Avelumab, Keytruda + Avelumab combination or isotype control antibodies.
 - ****, P<0.0001, **, P<0.01, *, P<0.05, Two-way ANOVA and Tukey's multiple comparisons test.

CTX-8371 is effective against human xenograft tumors

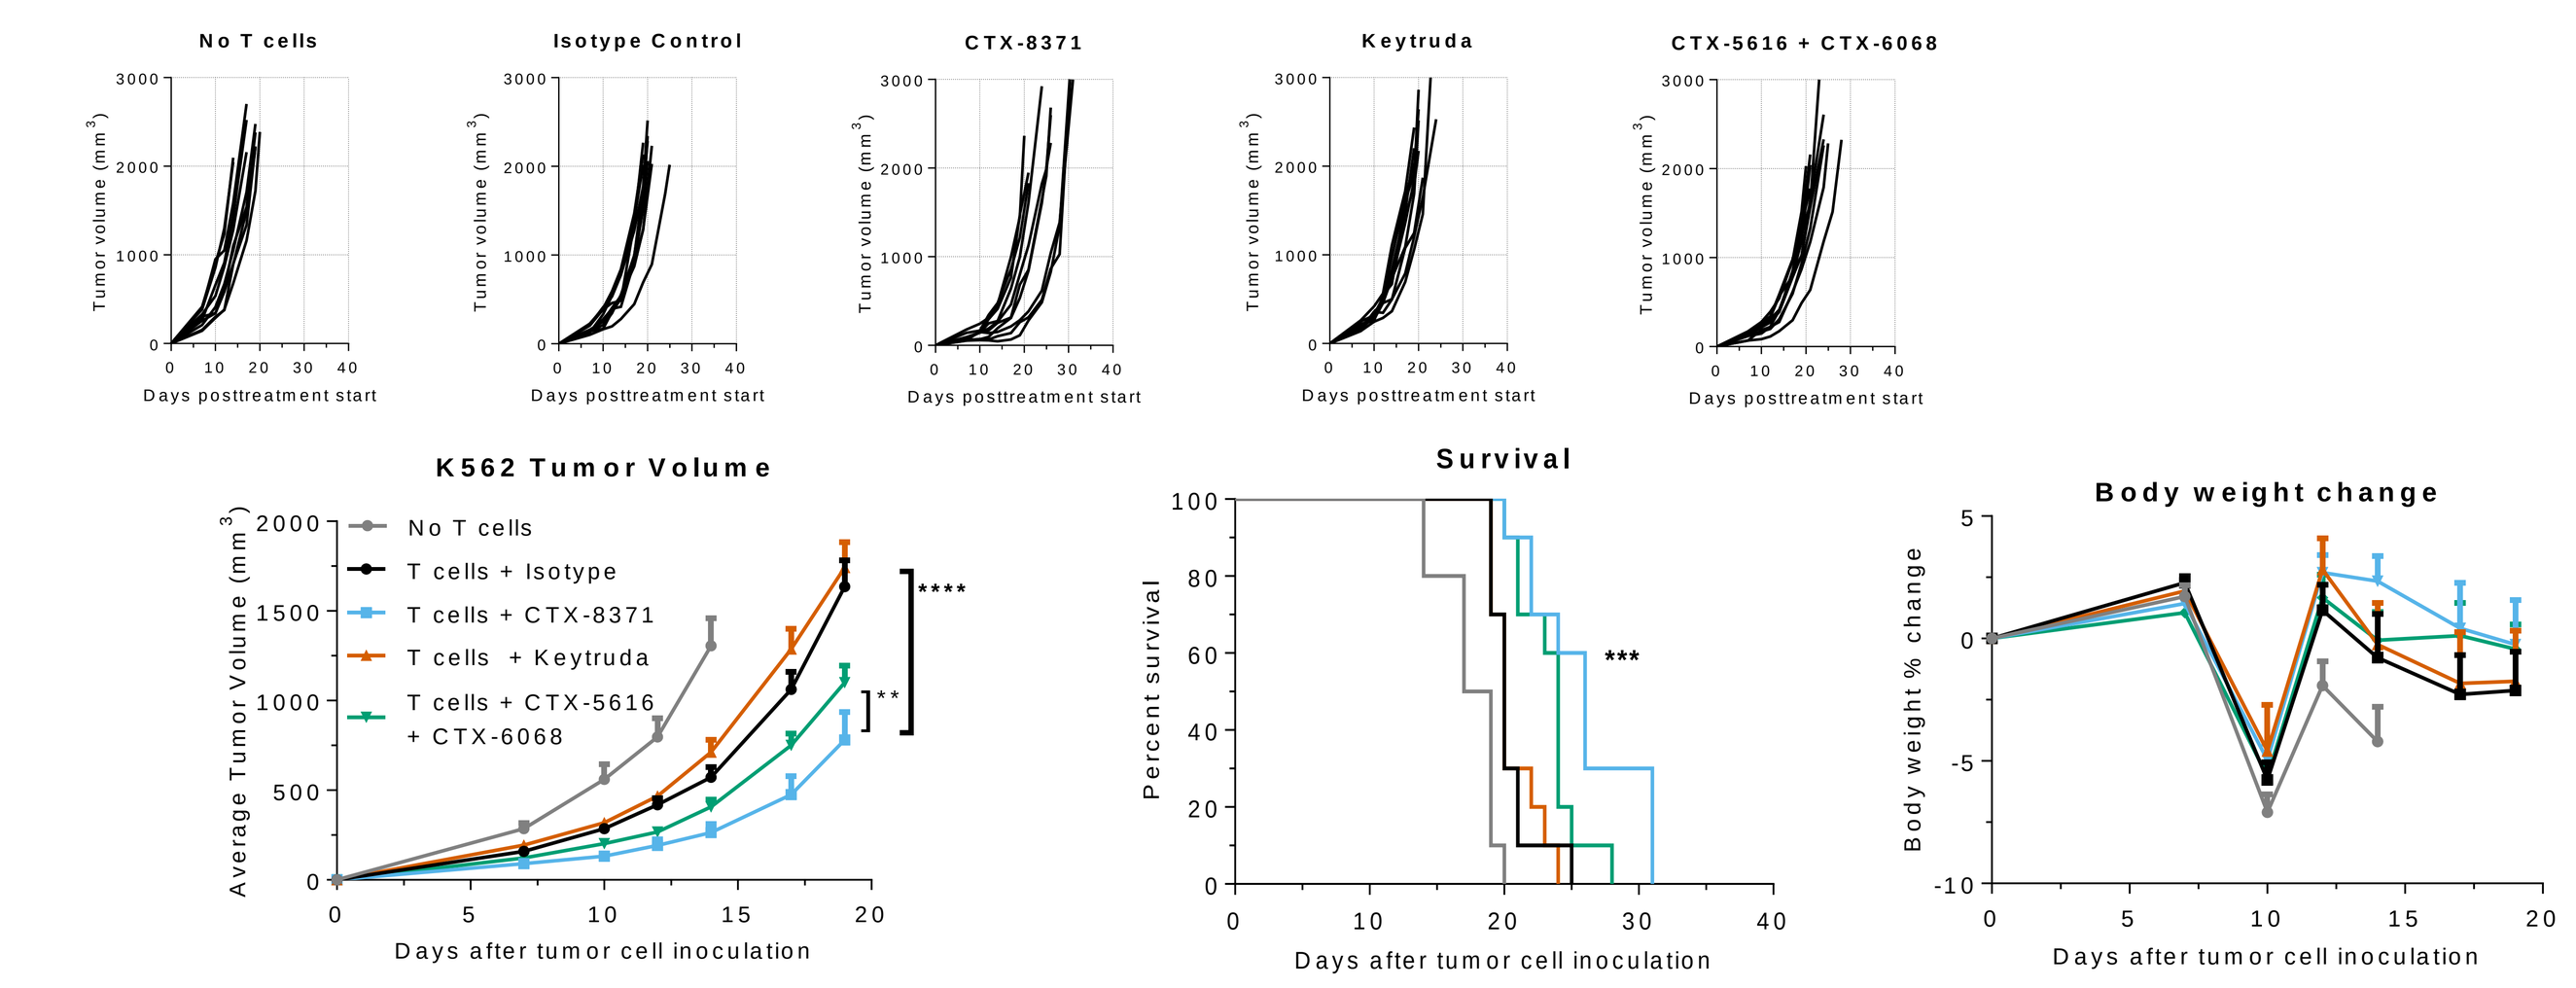
No T cells
Isotype Control
CTX-8371
Keytruda
CTX-5616 + CTX-6068
Days posttreatment start
K562 Tumor Volume
T cells + Isotype
T cells + CTX-8371
T cells + Keytruda
T cells + CTX-5616 + CTX-6068
Average Tumor Volume (mm3)
Survival
Percent survival
Body weight change
Body weight % change
Days after tumor cell inoculation

- NSG Mice were co-inoculated s.c. with 5e6 K562-A2-CMV-PDL1 and 5e6 CMV-specific T cells (upper panels) or 5e6 Raji-A2-CMV-PDL1 and 5e6 CMV-specific T cells (lower panels) in matrigel
- Dosing with equimolar amounts of antibody started at day of inoculation and every 3 days thereafter for a total of 5 doses
- Tumor size and body weight were measured 2-3 times per week, with mice euthanized when tumors are approaching 2000 mm^3 or mice lost 20% of body weight

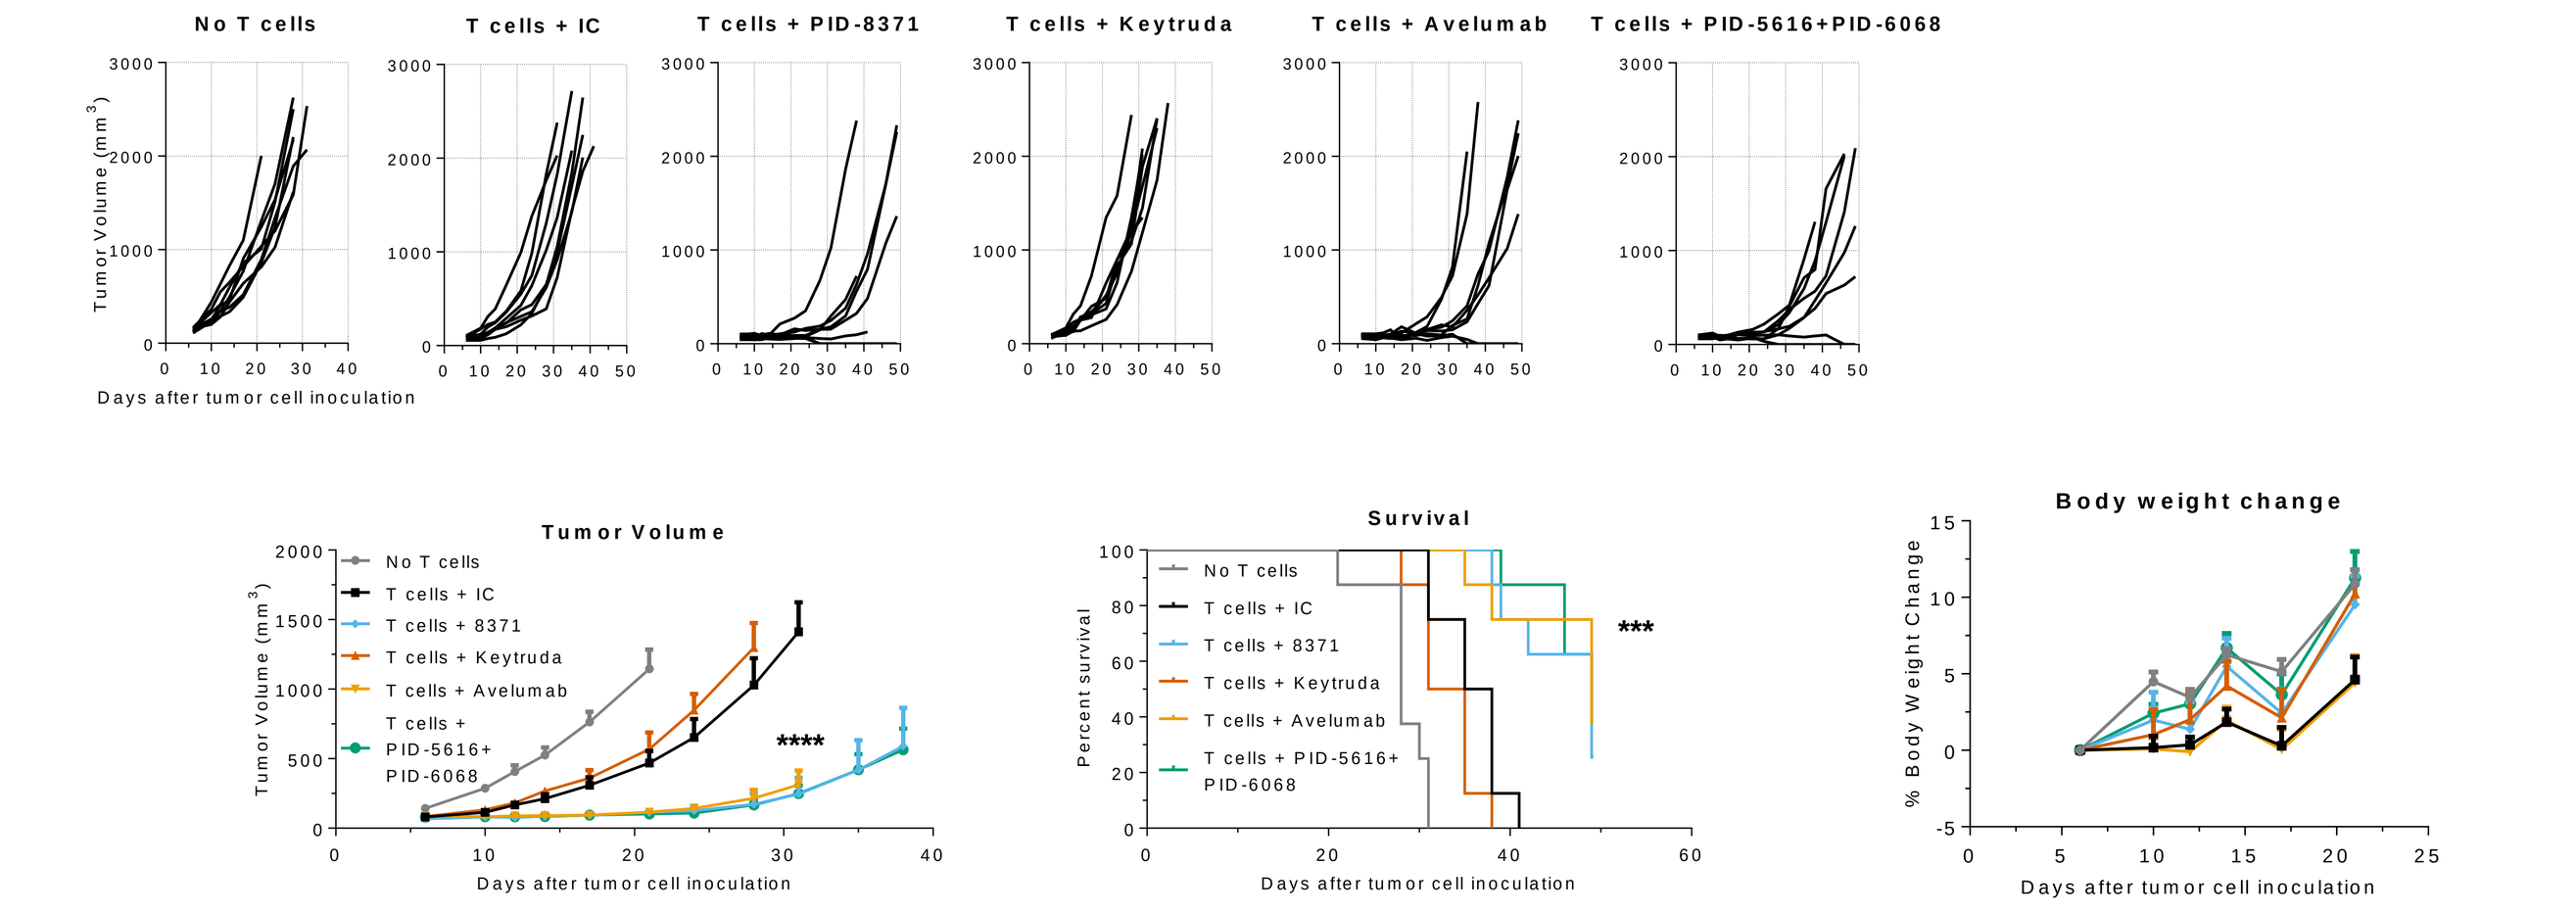
No T cells
T cells + IC
T cells + PID-8371
T cells + Keytruda
T cells + Avelumab
T cells + PID-5616+PID-6068
Tumor Volume
Survival
Body weight change
Days after tumor cell inoculation

Treatment	Tumor-free/Total
No T cells	0/8
T cells + IC	0/8
T cells + CTX-8371	2/8
T cells + Keytruda	0/8
T cells + Avelumab	2/8
T cells + a-PD-1 + a-PD-L1	2/8

- CTX-8371 is effective against human xenograft tumors
 - Upper panel shows individual, average tumor growth curves, survival, and body weight change of NSG mice adoptively transferred with CMV-specific human T cells against K562 s.c. tumor challenge. Different groups of mice (n=10) were treated with CTX-8371, Keytruda, anti-PD-1 + anti-PD-L1 monoclonal combination or isotype control antibodies.
 - Lower panel shows individual, average tumor growth curves, survival, and body weight change of NSG mice adoptively transferred with CMV-specific human T cells against Raji s.c. tumor challenge. Different groups of mice (n=8) were treated with CTX-8371, Keytruda, Avelumab, anti-PD-1 + anti-PD-L1 monoclonal combination or isotype control antibodies.
 - ****, P<0.0001, **, P<0.01, Two-way ANOVA and Tukey's multiple comparisons test; ***, *P*<0.001, Log-rank (Mantel-Cox) test comparing CTX-8371 to Keytruda .

Summary

- PD-1xPD-L1 Stichmab™ antibodies were superior to monoclonal combination in a MLR assay. This preliminary data led to the creation of CTX-8371, a common light chain bispecific antibody with excellent drug-like properties.
- CTX-8371 had exceptional *in vitro* activity and *in vivo* anti-tumor efficacy compared to approved checkpoint inhibitor immunotherapies and combination thereof.
- The anti-tumor activity of CTX-8371 might be at least in part explained by its unique MOA leading to massive surface PD-1 loss, mediated through bridging the T cell and tumor cell.
- Investigation into CTX-8371 MOA is ongoing.